Contrast-modulated noise shows an adaptable, rectifying, contrast-comparison process ("Buffy adaptation")

S. Sabina Wolfson, Stephanie Pan, Gauri Wable & Norma Graham

Columbia University, Department of Psychology, New York, NY

» Introduction «

We found a contrast-comparison process that *adapts* (to the recent average contrast) and *rectifies* (on the contrast – not luminance – dimension).

Previously we have demonstrated this process using patterns composed of regularly spaced Gabor patch elements (Wolfson & Graham, *JOV*, 2007).

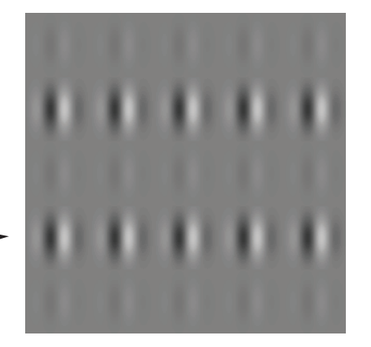

Here we use contrast-modulated noise patterns.

In both of these cases, we find that performance is poor on test patterns with contrasts that STRADDLE the adapt contrast (that is, one test contrast is higher and the other is lower). Performance is better on test patterns with an average test contrast a bit ABOVE or BELOW the adapt contrast.

We call this the Straddle Effect.

» Psychophysical trial «

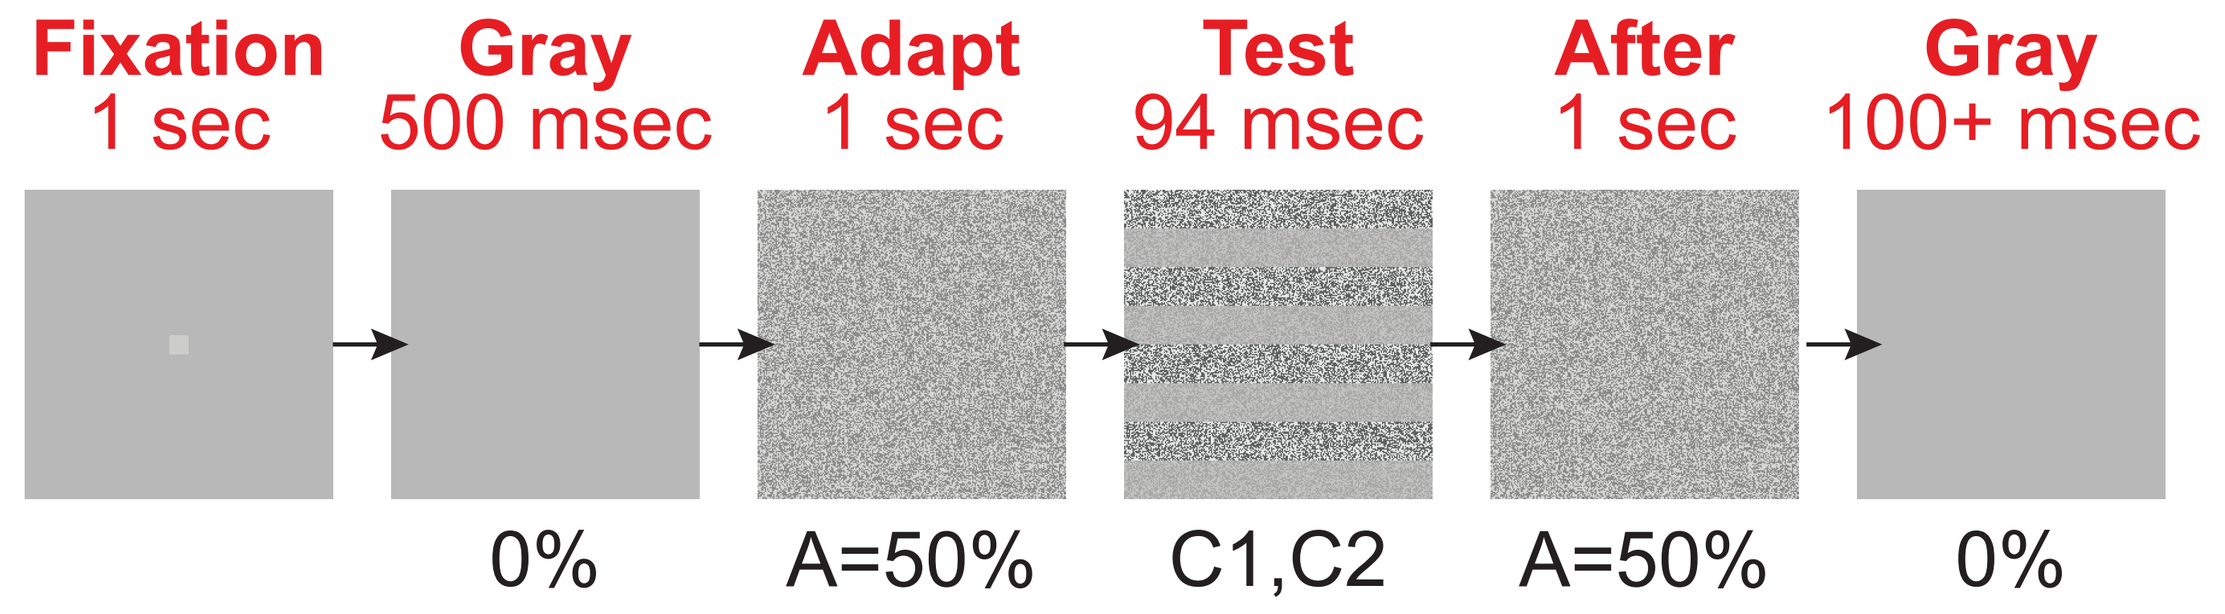
Fixation
1 sec
Gray
500 msec
Adapt
1 sec
Test
94 msec
After
1 sec
Gray
100+ msec
0%
A=50%
C1,C2
A=50%
0%

A – the adapt contrast – is always 50%
C1,C2 – the two contrasts in a test pattern
C_{avg} – the average test contrast – is (C1+C2)÷2
ΔC – the test contrast difference – is |C1-C2|
C1 and C2 are symmetric around C_{avg} so
$C1 = C_{avg} + \frac{1}{2}\Delta C$ and $C2 = C_{avg} - \frac{1}{2}\Delta C$

» Task «

Identify orientation of test pattern's contrast-defined stripes.

"vertical" 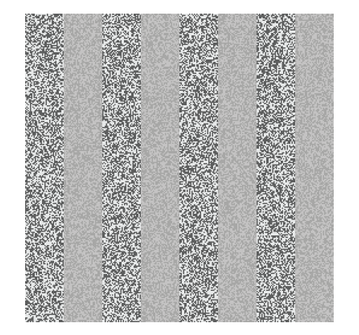 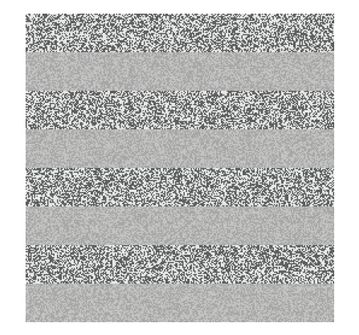 "horizontal"

» Methodological details «

- Observers were paid Columbia University undergraduates with normal or corrected-to-normal vision.
- Feedback was provided.
- Patterns were 16 x 16 deg at the viewing distance of ~0.9m.
- The contrast-defined stripe periodicity was 0.5 cyc/deg.
- The mean luminance was approx 50 cd/m^2.
- Each noise check was 2x2 pixels (64 pixels = 1 deg).
- The luminance of each noise check was drawn from a binary (or uniform) distribution.

» Noise THRESHOLD Results «

Sharp-edged contrast-defined stripes

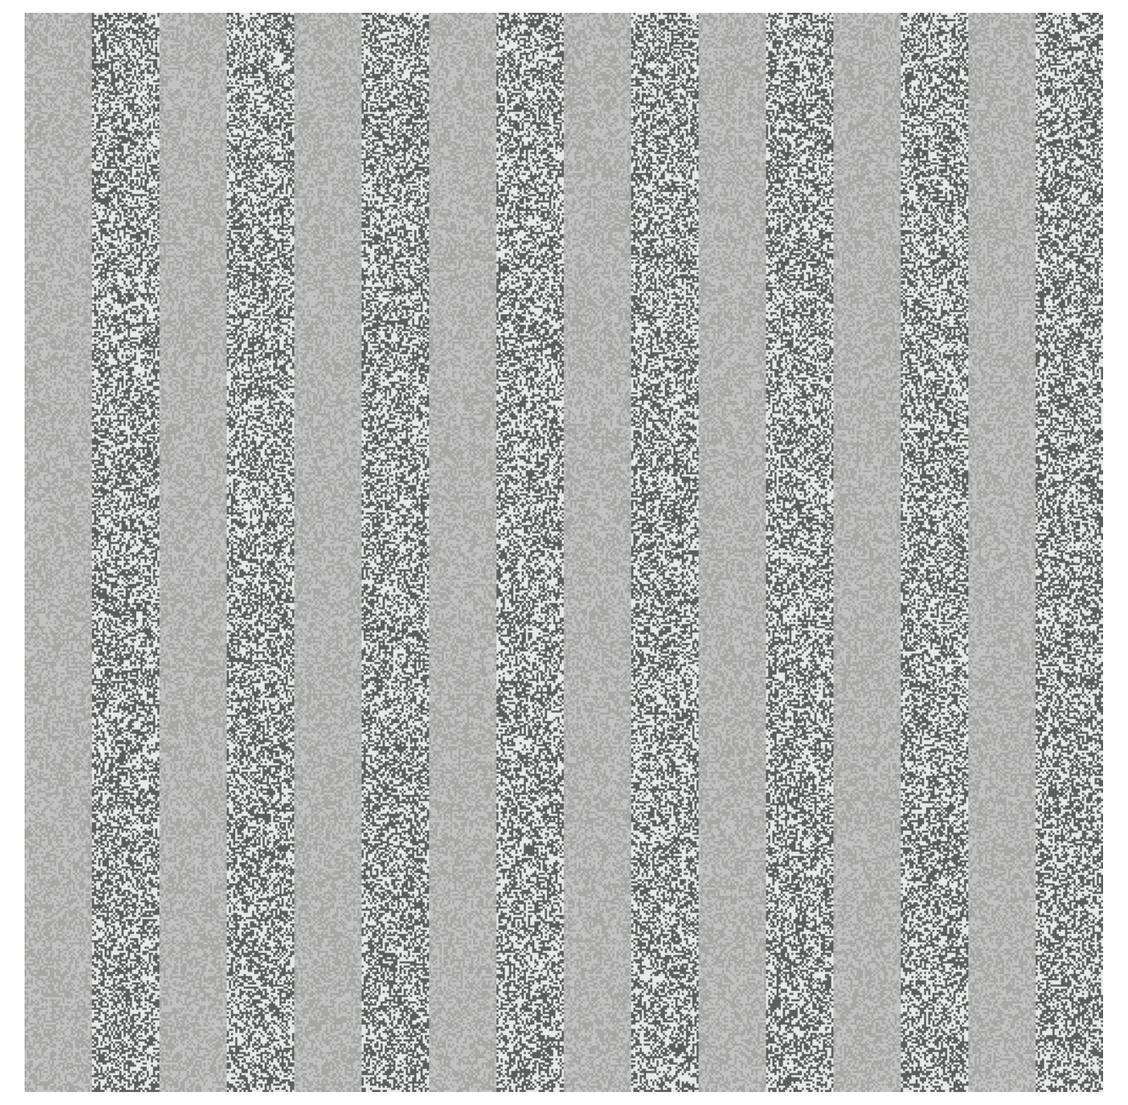

Square-element contrast-defined stripes

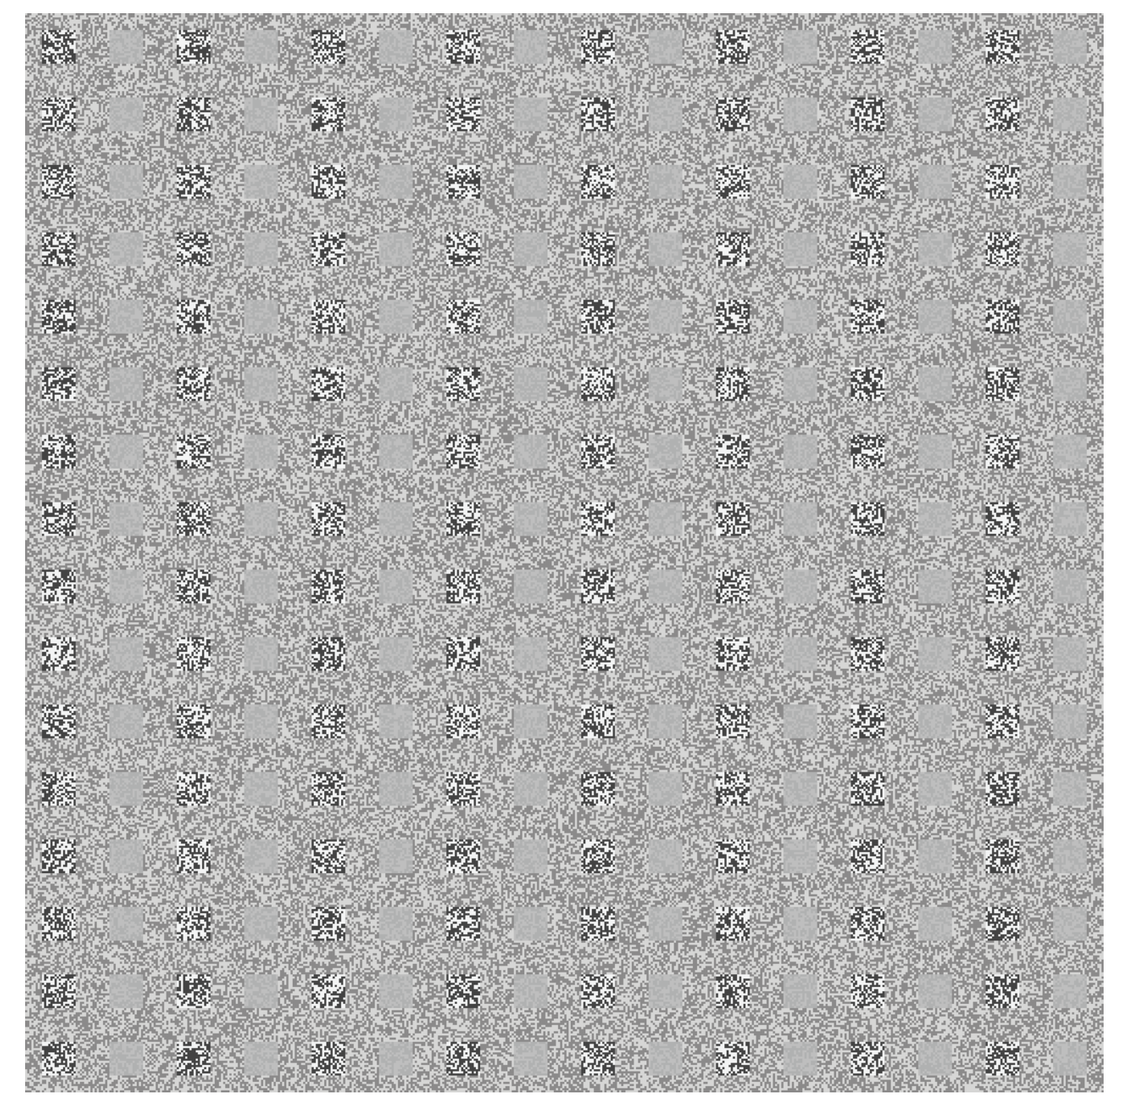

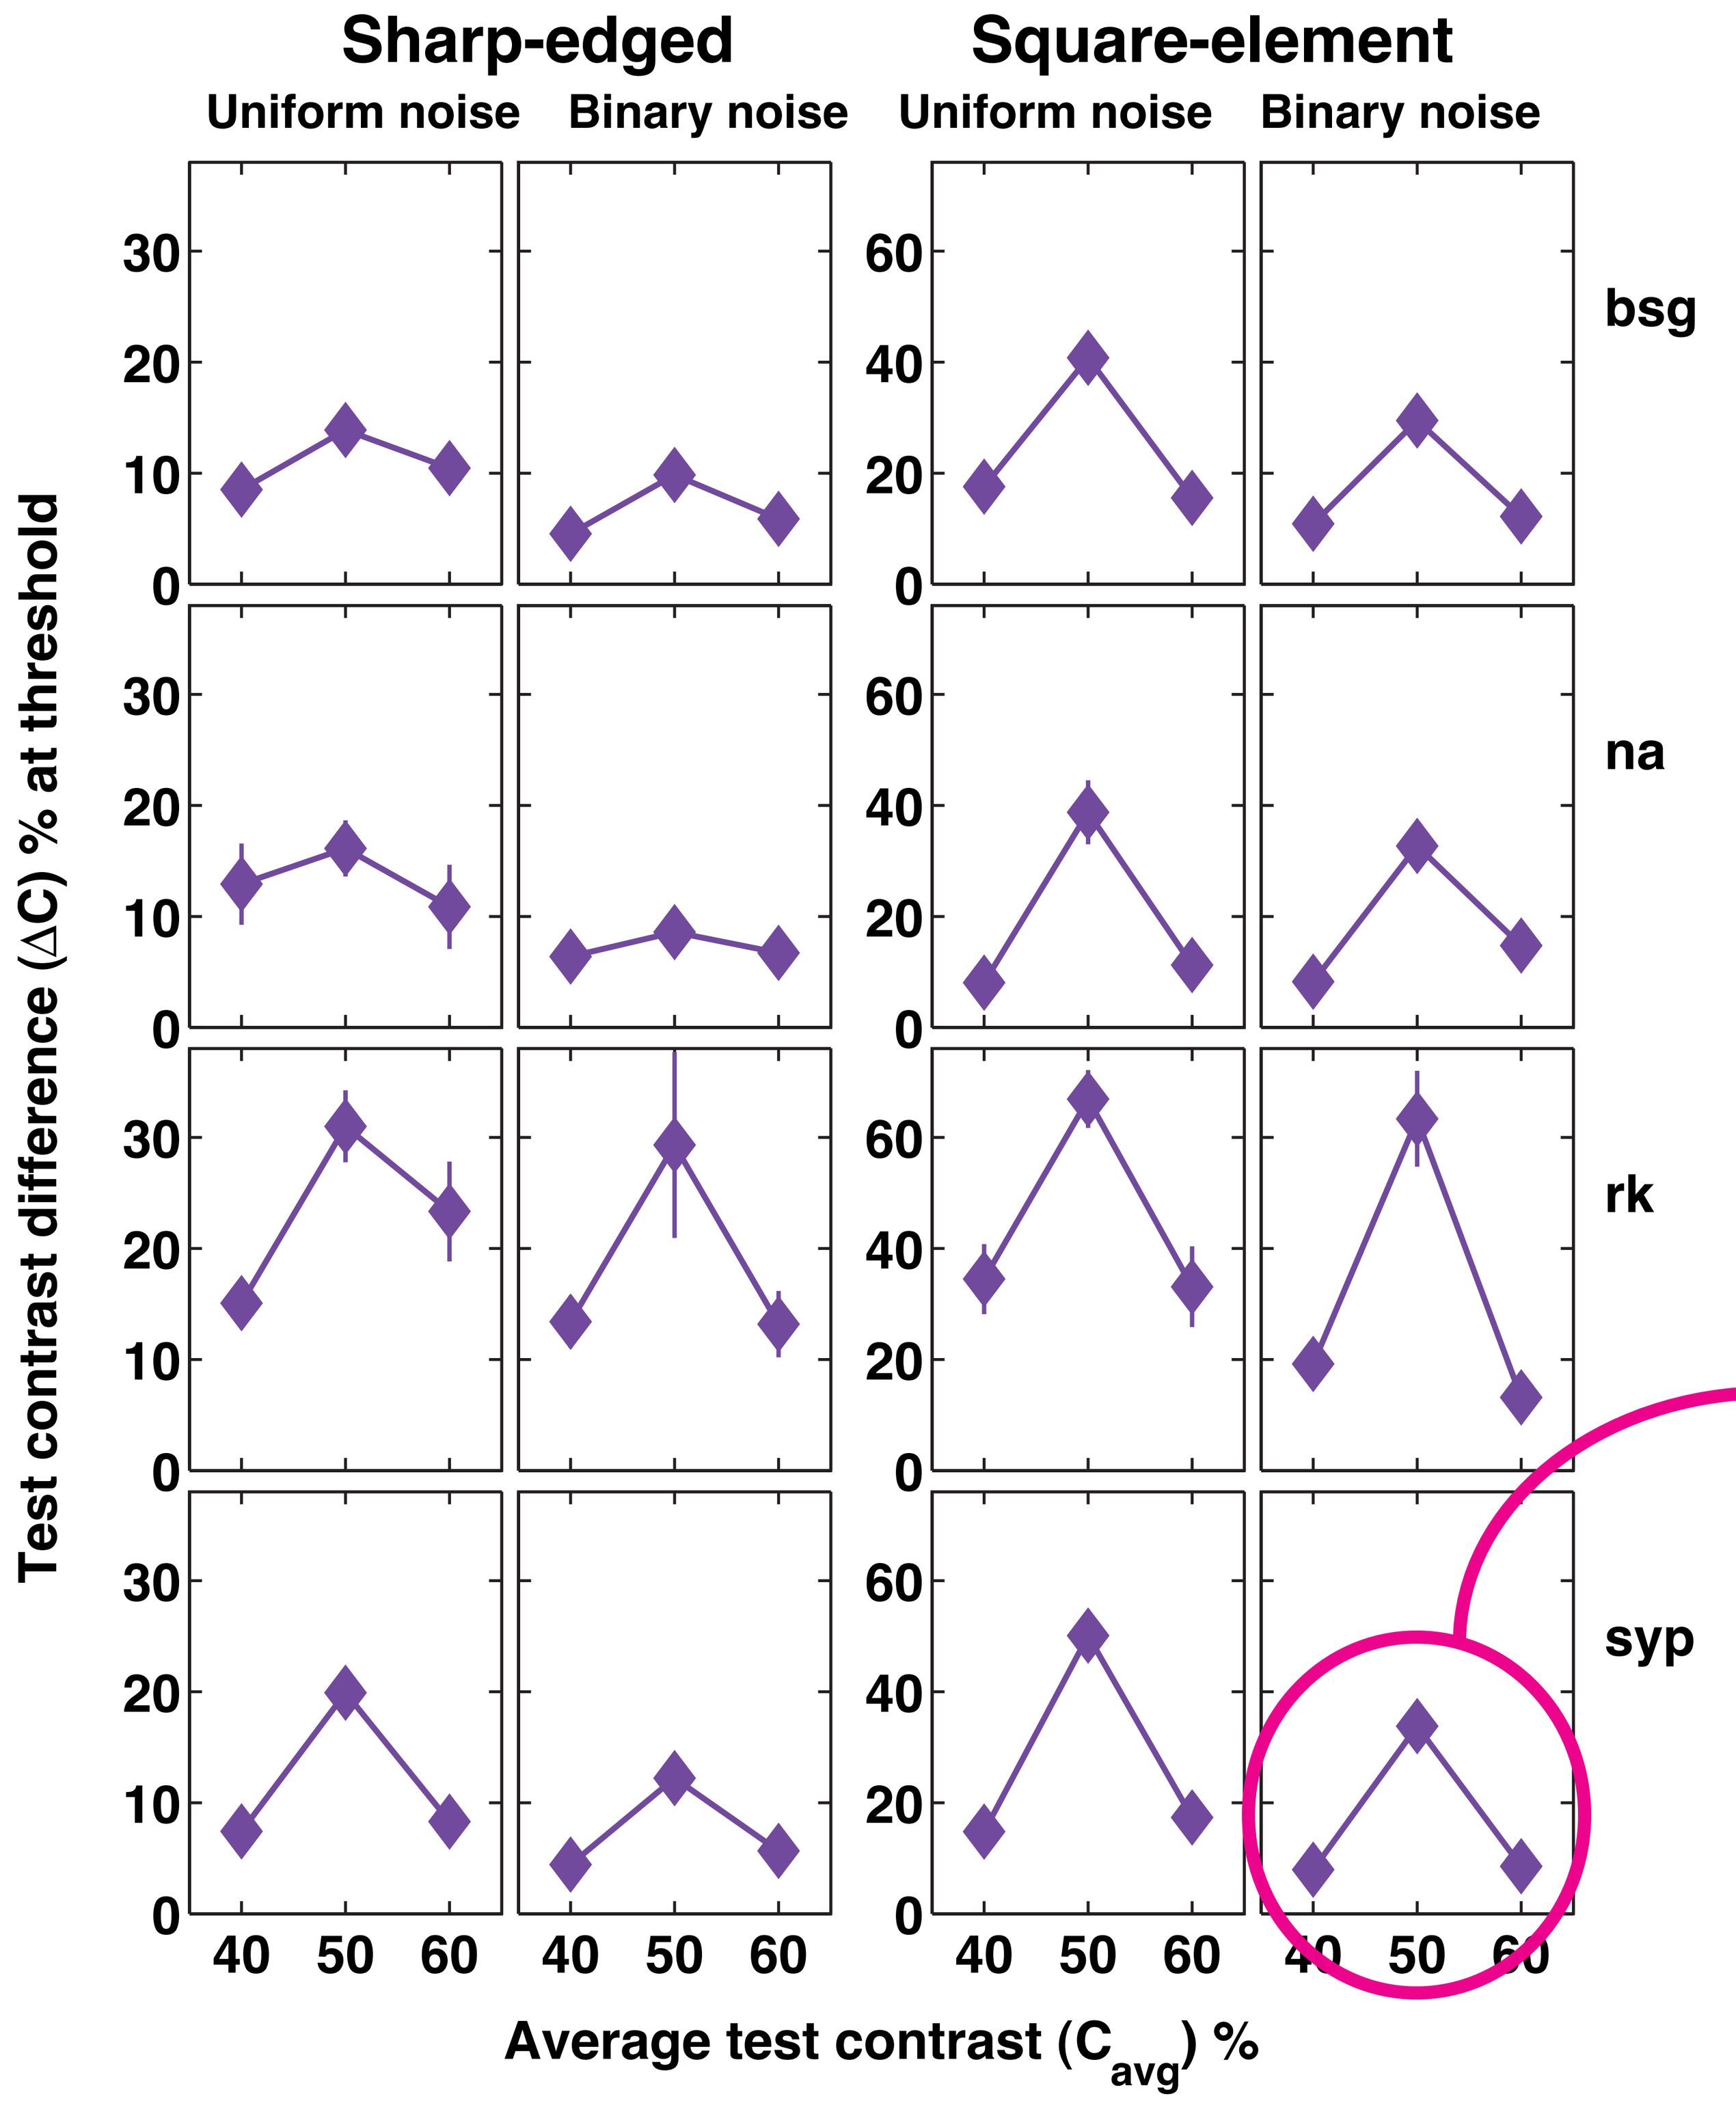
Sharp-edged
Square-element
Uniform noise
Binary noise
Uniform noise
Binary noise
bsg
na
rk
syp
Test contrast difference (ΔC) % at threshold
Average test contrast (Cavg) %

» THRESHOLD experiment methods «

For each average test contrast (C_{avg} = 40%, 50%, 60%), adjust test contrast difference (ΔC) in a staircase (3-up-1-down, 30 trials). Estimate (80% correct) threshold for each session (for each C_{avg}). Plotted here is the mean of the threshold estimates +or- 1 SE.

» CONSTANT DIFFERENCE Results «

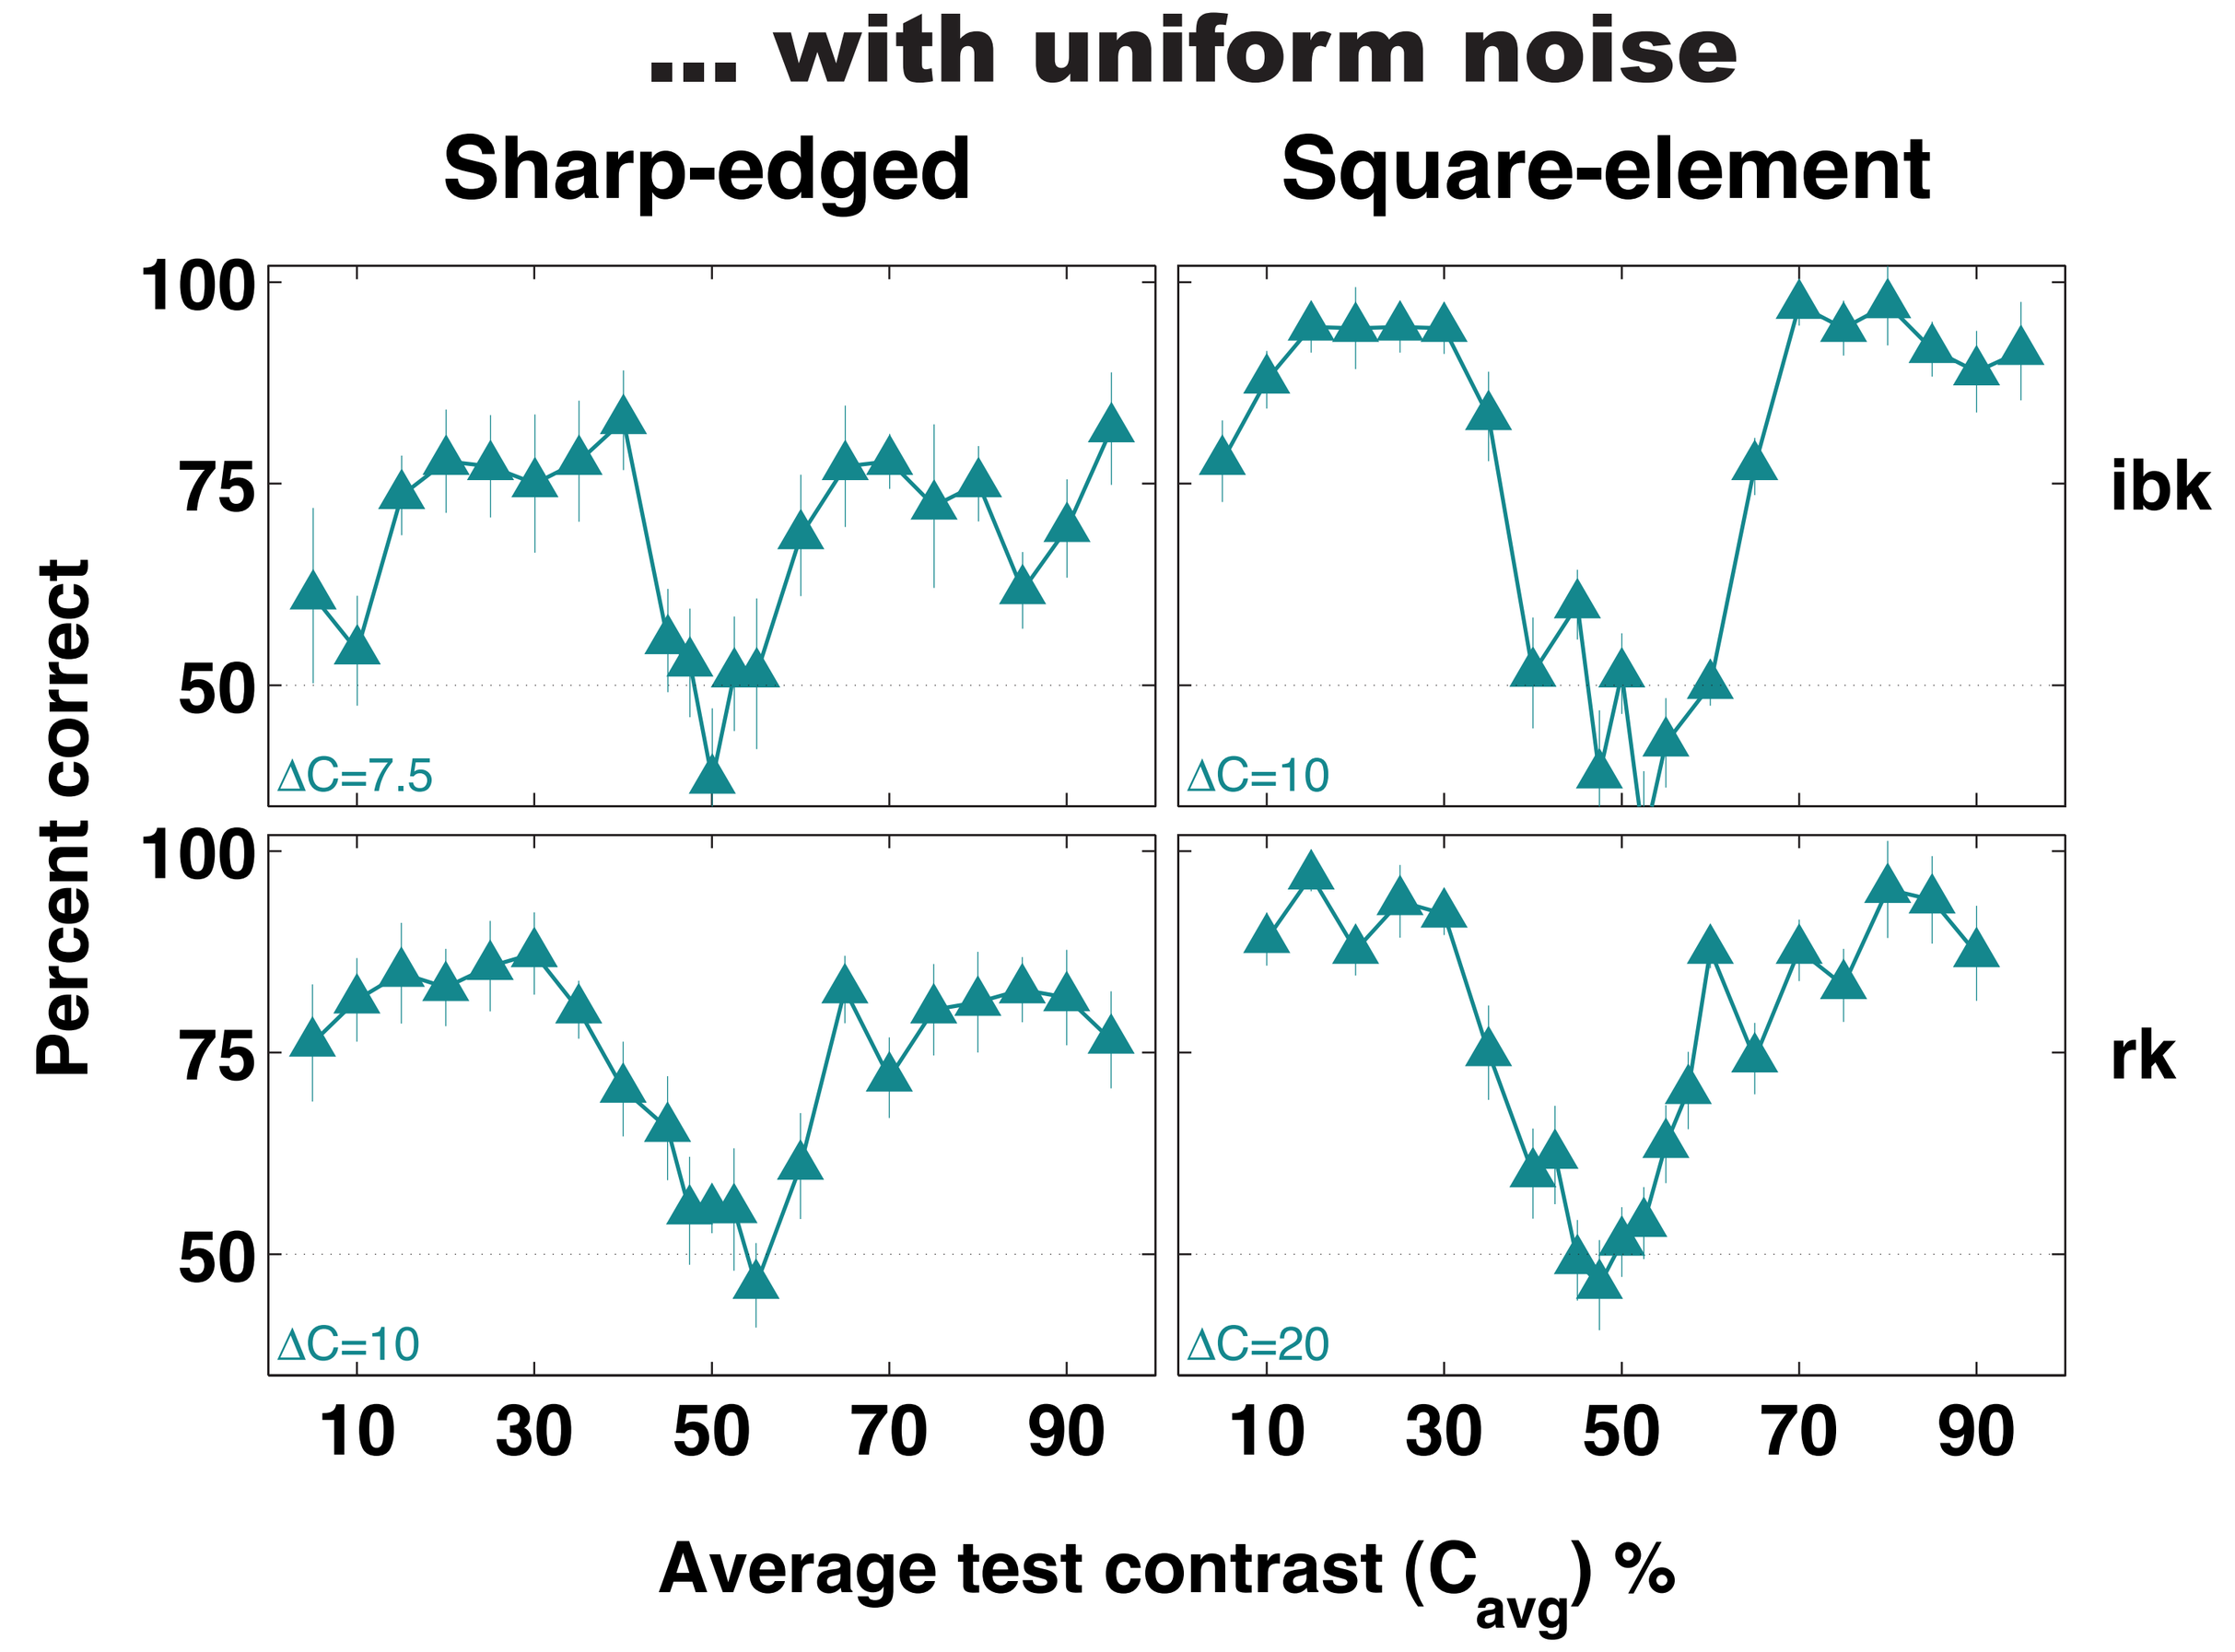
... with uniform noise
Sharp-edged
Square-element
ibk
rk
Percent correct
ΔC=7.5
ΔC=10
ΔC=10
ΔC=20
Average test contrast (Cavg) %

» CONSTANT-DIFFERENCE experiment methods «

For each average test contrast (C_{avg}), measure performance while holding the test contrast difference (ΔC) constant (7.5%, 10%, or 20% depending on observer and condition). Plotted here is percent correct identification +or- 1 SE.

A closer look at these three thresholds...

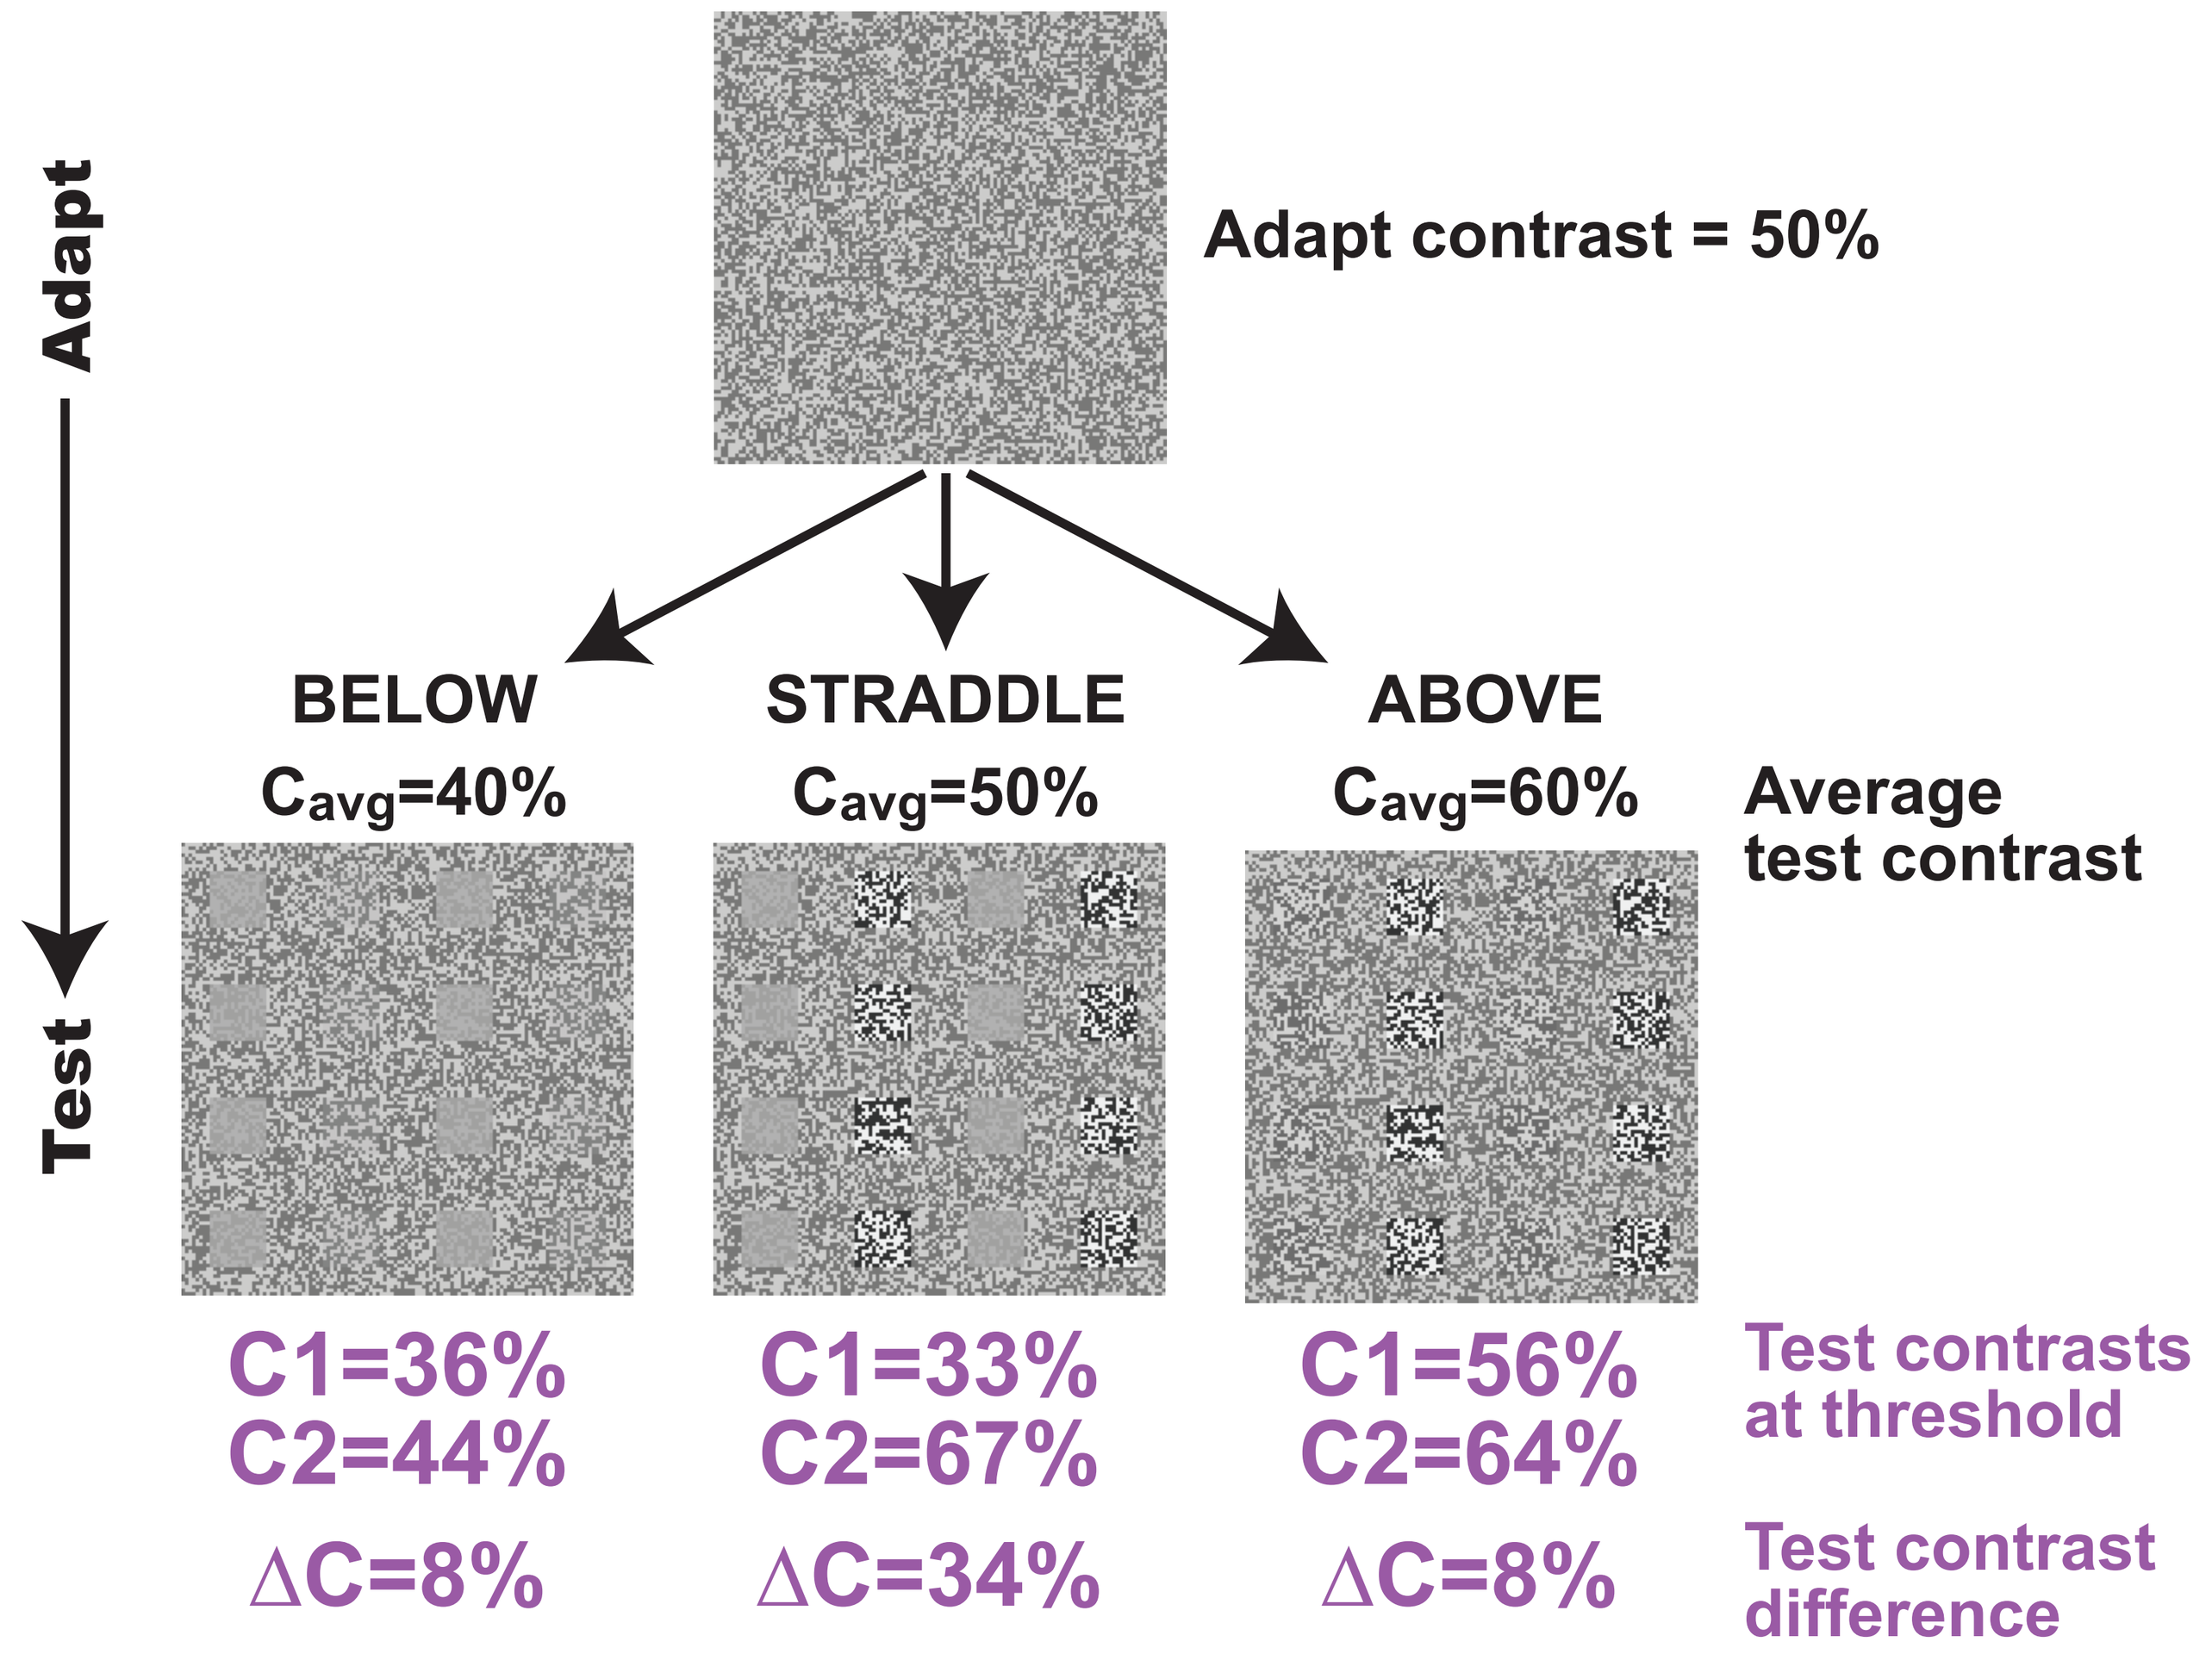
Adapt contrast = 50%
Adapt
Test
BELOW
Cavg=40%
STRADDLE
Cavg=50%
ABOVE
Cavg=60%
Average test contrast
C1=36%
C2=44%
C1=33%
C2=67%
C1=56%
C2=64%
Test contrasts at threshold
ΔC=8%
ΔC=34%
ΔC=8%
Test contrast difference

Performance on each of these test patterns is equal by definition, but the STRADDLE pattern's contrasts actually span the others!

This is a huge Straddle Effect.

» Brief aside on modelling «

To account for the poor performance on STRADDLE test patterns, we insert a *contrast comparison process* in a complex (second-order) channel.

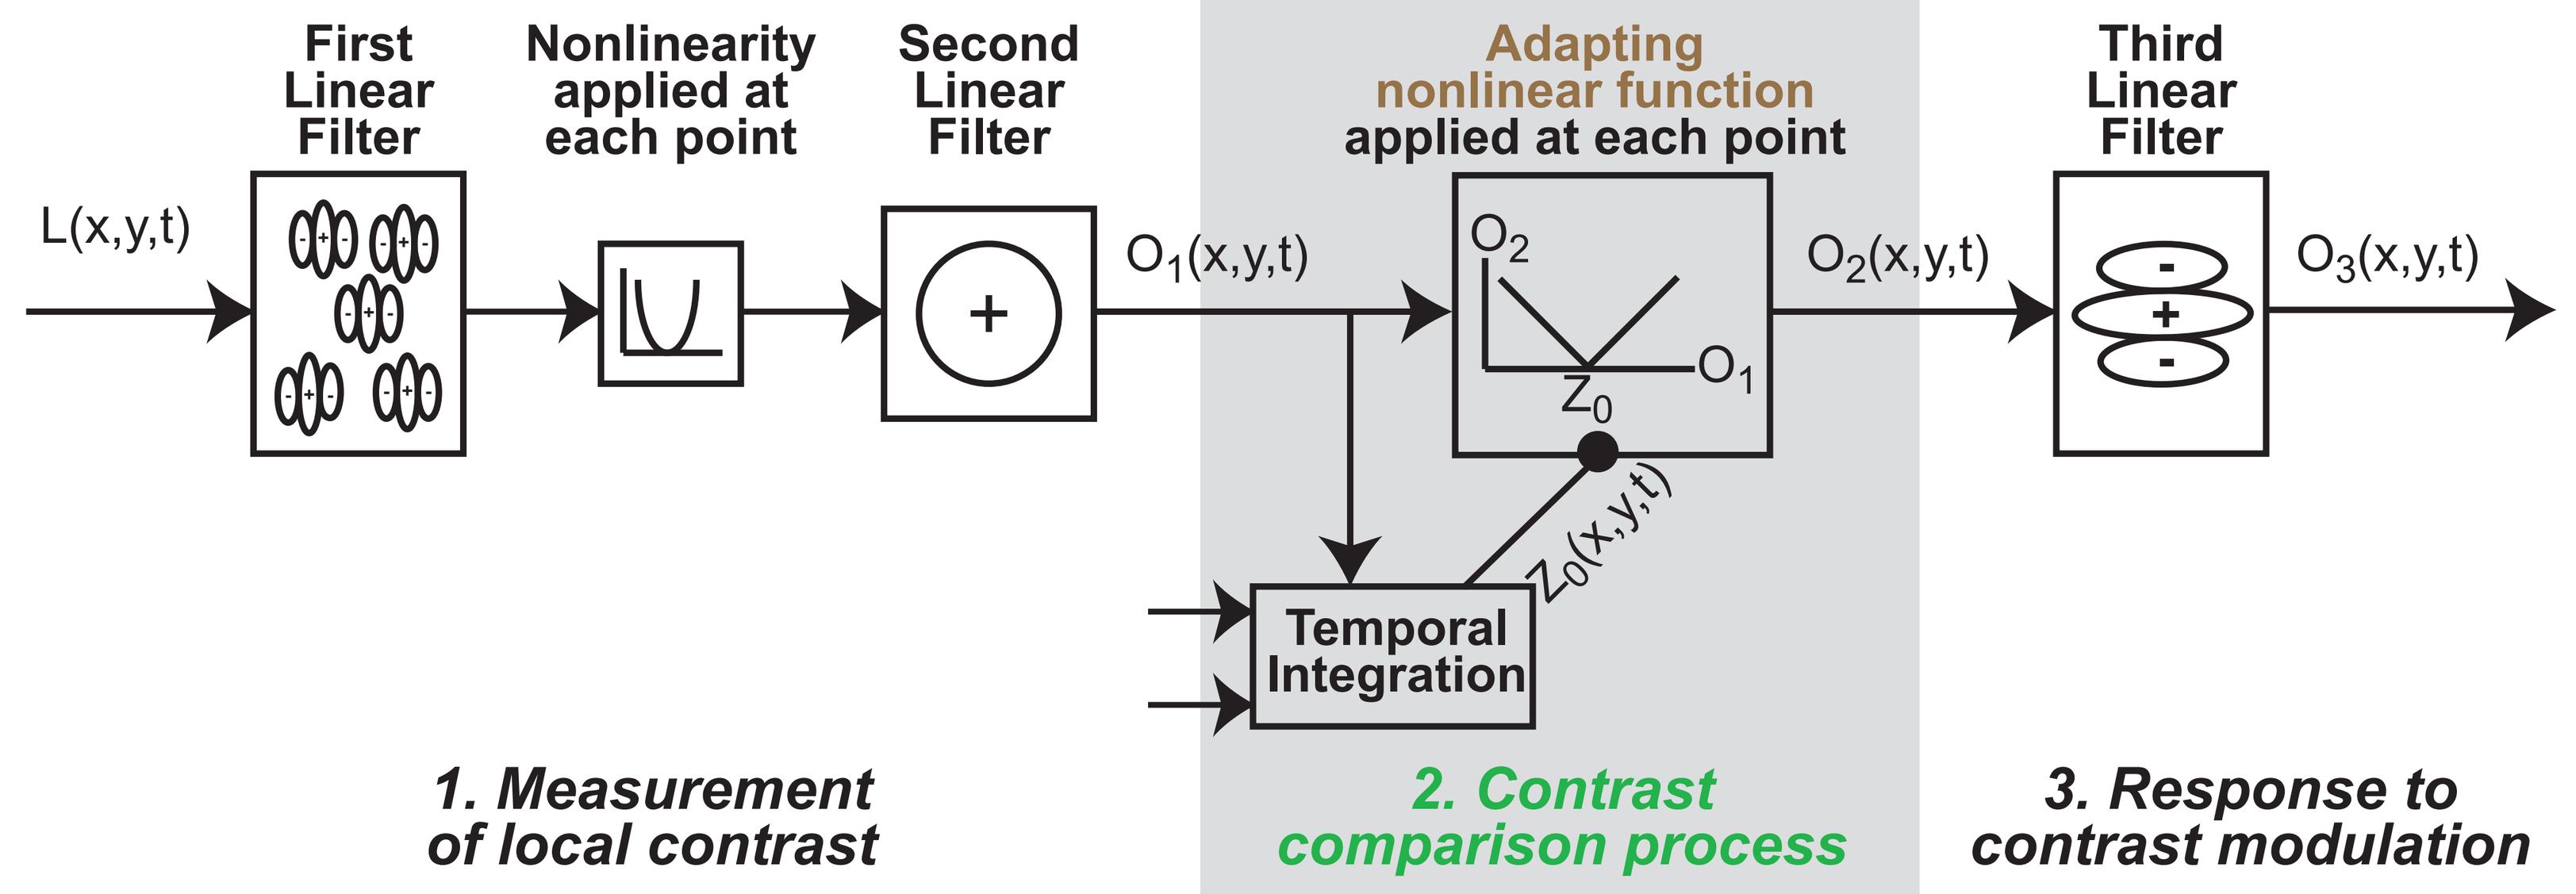
First Linear Filter
Nonlinearity applied at each point
Second Linear Filter
Adapting nonlinear function applied at each point
Third Linear Filter
L(x,y,t)
O1(x,y,t)
O2
O1
Z0
O2(x,y,t)
O3(x,y,t)
Z0(x,y,t)
Temporal Integration
1. Measurement of local contrast
2. Contrast comparison process
3. Response to contrast modulation

The *adapting nonlinear function* within the *contrast comparison process* rectifies on the contrast dimension (not the luminance dimension) and its zero-point changes (adapts) based on the recent average contrast.

If the rectification were full-wave as shown, all STRADDLE patterns would be invisible to the channel. Instead, we use a pair of (otherwise identical) channels, one of which prefers contrast increases ("*On*") and the other decreases ("*Off*").

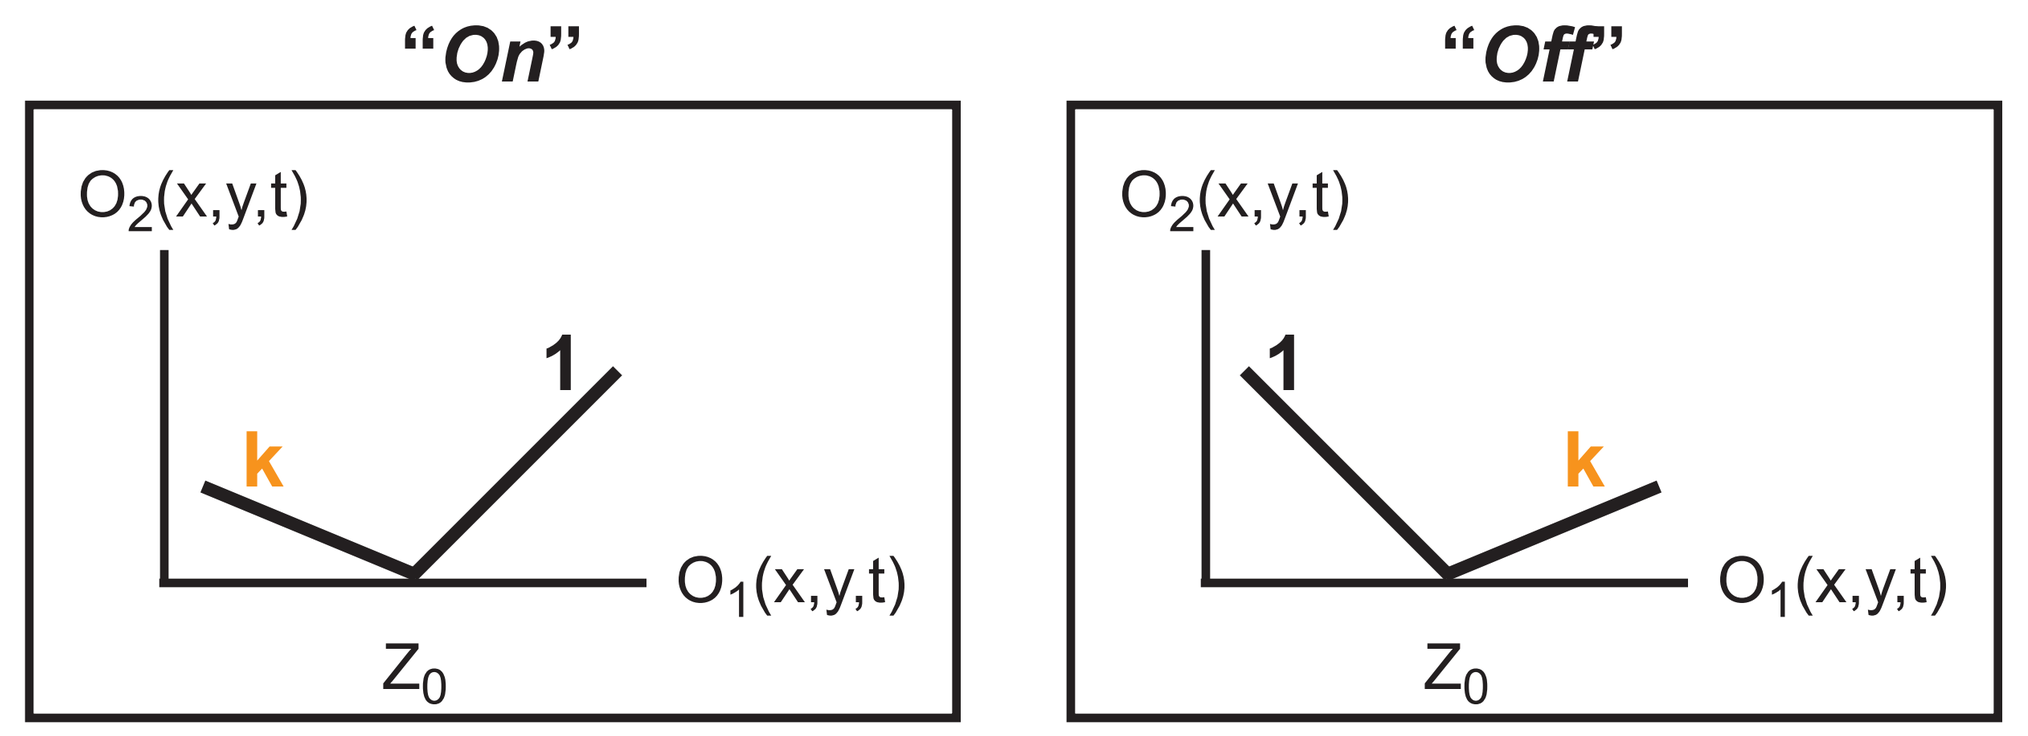
"On"
"Off"
O2(x,y,t)
k
1
O1(x,y,t)
Z0
O2(x,y,t)
1
k
O1(x,y,t)
Z0

k is the ratio of the slopes (k = 0 for half-wave, k=1 for full-wave).

The value of k is determined by an observer's STRADDLE & Non-STRADDLE thresholds.

This model explains the Straddle Effect. To explain the full range of results, contrast normalization must be added to the model (Wolfson & Graham, *JOV*, 2009).

» Conclusions «

We find poor performance on STRADDLE test patterns relative to test patterns a bit ABOVE and BELOW (the adapt contrast).

This Straddle Effect is bigger with the square-element test patterns than the sharp-edged test patterns.

Since we have now found the Straddle Effect for noise patterns as well as grids of regularly spaced Gabor patches, we suspect that this *contrast comparison process* has to be in any model of contrast processing.

A possibility: In most situations, it isn't raw contrast that counts. Instead, it is something more like the (unsigned) difference between the current contrast and the recent average contrast.

www.columbia.edu/~sw354
www.columbia.edu/~nvg1
VSS 2009

References:

Wolfson, S. S. & Graham, N. (2007). An unusual kind of contrast adaptation: Shifting a contrast comparison level. *Journal of Vision*, Vol 7, Num 8, Article 12. http://journalofvision.org/7/8/12/

Wolfson, S. S. & Graham, N. (2009). Two contrast adaptation processes: Contrast normalization and shifting, rectifying contrast comparison. *Journal of Vision*, Vol 9, Num 4, Article 30. http://journalofvision.org/9/4/30/